Focus on Controlling Non-peptide Impurities in Peptide API Manufacturing:

Detection, Identification and Removal of a DTT-di-*t*-Bu Adduct Formed in Peptide Resin TFA Cleavages

Jan Pawlas,[1] Stefan Hansen, Thomas Svensson and Gunnar Stærkær

PolyPeptide Laboratories (Sweden) AB, Limhamnsvägen 108, PO BOX 30089, 20061 Limhamn, Sweden

Introduction

In contemporary peptide API manufacturing considerable resources are devoted to development of analytical methods suitable for the detection of impurities.[2] While these methods are typically quite adequate for the detection of peptide-related impurities, they may not be equally suited for the detection of minute amounts of low MW non-peptide entities. Nevertheless, small molecule reagents, reactants, solvents as well as amino acid protecting groups can be encountered throughout any peptide API manufacturing process. Incomplete removal of such compounds and/or formation of new small molecules during the manufacturing can compromise the quality of a peptide API. In fact, we recently reported that the reducing agent DTT (Cleland's reagent, **1** in Figure 1)[3-4] can form such a difficult-to-detect/remove impurity (dithioorthoester **2**, Figure 1) during peptide resin TFA cleavages.[5] We now report that employing DTT as a scavenger in TFA cleavages can cause the formation of another cumbersome non-peptide species (dithioether **3**, Figure 1). Means of removing **3** during downstream processing of a 31-mer peptide API is also discussed herein.

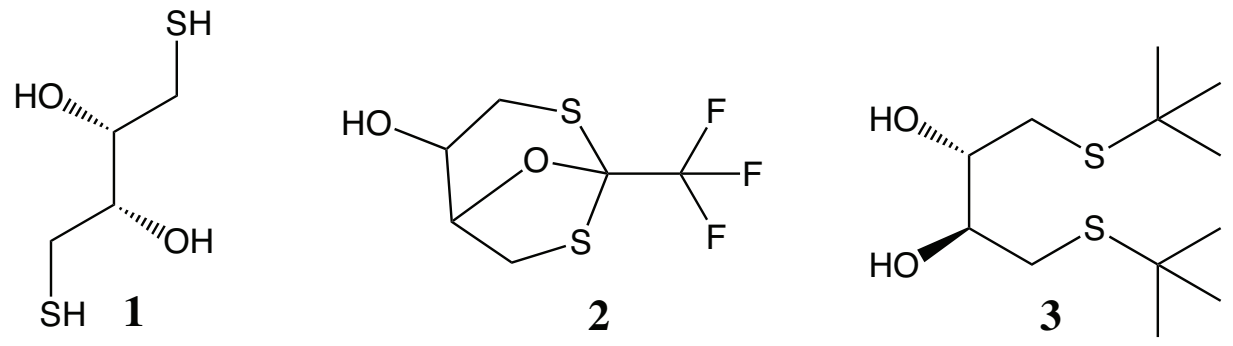
SH
HO
OH
SH
1
HO
O
S
S
F
F
F
2
HO
S
HO
S
3

Figure 1. DTT (**1**) and its adducts with TFA (**2**) and 2 x *t*-Bu (**3**), respectively.

Results & Discussion

During a recent Fmoc-SPPS manufacturing campaign of a 31-mer peptide amide we encountered a low MW (< 300) impurity in crude solutions of the peptide. In analytical HPLC systems this low MW compound eluted quite far from the main peak (Rrt ~ 0.25) albeit it did exhibit a propensity to co-elute with the product during our initial preparative RP-HPLC experiments. In keeping with its UV spectrum (Figure 2), this impurity was poorly detectable at ≥ 220 nm but its absorption increased significantly at ≤ 210 nm.

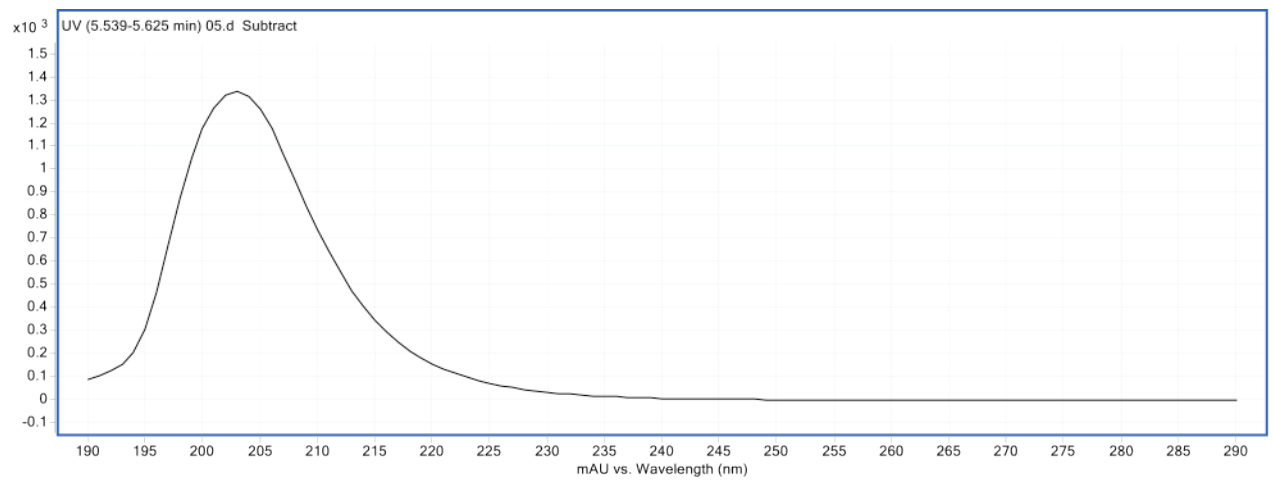
UV (5.539-5.625 min) 05.d Subtract
x10 3
mAU vs. Wavelength (nm)

Figure 2. UV spectrum of a low MW impurity detected in the solutions of a 31-mer peptide API.

We isolated this material from the peptide solutions at hand as a 99.4% pure white crystalline solid,[6] elucidated it by HRMS (Figure 3), elemental analysis[7] and NMR[8-9] and determined its structure to be the DTT-di-*t*-Bu adduct **3** (Figure 1). The structure of **3** suggests that it was formed during the TFA cleavage/global deprotection of the 31-mer peptide resin by a reaction of DTT with *t*-Bu cations released from the side chain protecting groups.

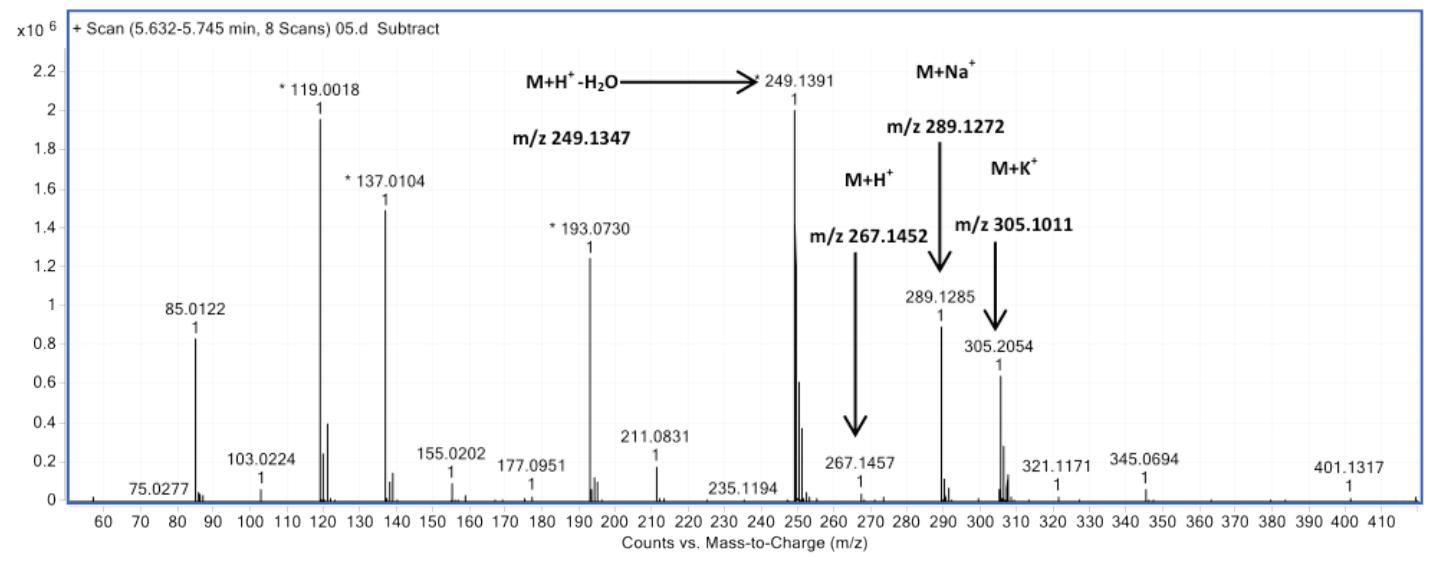
+ Scan (5.632-5.745 min, 8 Scans) 05.d Subtract
x10 6
M+H+-H2O
m/z 249.1347
M+Na+
m/z 289.1272
M+H+
m/z 267.1452
M+K+
m/z 305.1011
249.1391
119.0018
137.0104
193.0730
85.0122
289.1285
305.2054
267.1457
211.0831
155.0202
177.0951
103.0224
75.0277
235.1194
321.1171
345.0694
401.1317
Counts vs. Mass-to-Charge (m/z)

Figure 3. HRMS spectrum of **3**.

In fact, HPLC quantifications of the adduct **3** produced in the aforementioned cleavage revealed that ~ 80 % of all *t*-Bu groups present on the resin reacted with DTT to form **3**, which shows that DTT is not only a good reducing agent but an excellent *t*-Bu$^+$ scavenger as well. It is worth noting that depending on the TFA cleavage conditions/characteristics of the peptide resin DTT can form either the bicyclic TFA adduct **2** and/or the di-*t*-Bu adduct **3**. For example, reacting TFA with DTT in the presence of TIS afforded the TFA adduct **2** essentially quantitatively.[10] On the other hand, reacting TFA with DTT in the presence of the *t*-Bu containing MTBE gave the di-*t*-Bu adduct **3** as the main product, and only a small amount of the TFA adduct **2** was formed.

Finally, although the di-*t*-Bu impurity **3** could be removed from the aq. API solutions fairly easily for example by an extraction with a suitable organic solvent, we set out to develop a more peptide manufacturing amenable RP-HPLC based protocol for the removal of **3**. Towards this end, we initially carried out test purifications using aq. TFA/MeCN buffers, which in analytical HPLC systems gave excellent product vs **3** separations. Interestingly, all purifications aiming at removing **3** using aq. TFA/MeCN mobile phases resulted in substantial peptide API (MW >3000) & adduct **3** (MW 266) co-elutions, regardless of the stationary phase used. Nevertheless, after some experimentation we found that simply using phosphate buffer pH 7.5/MeCN mobile phases facilitated adduct **3** removals without any appreciable product loss on every stationary phase that we examined (for an example, see Figure 4).

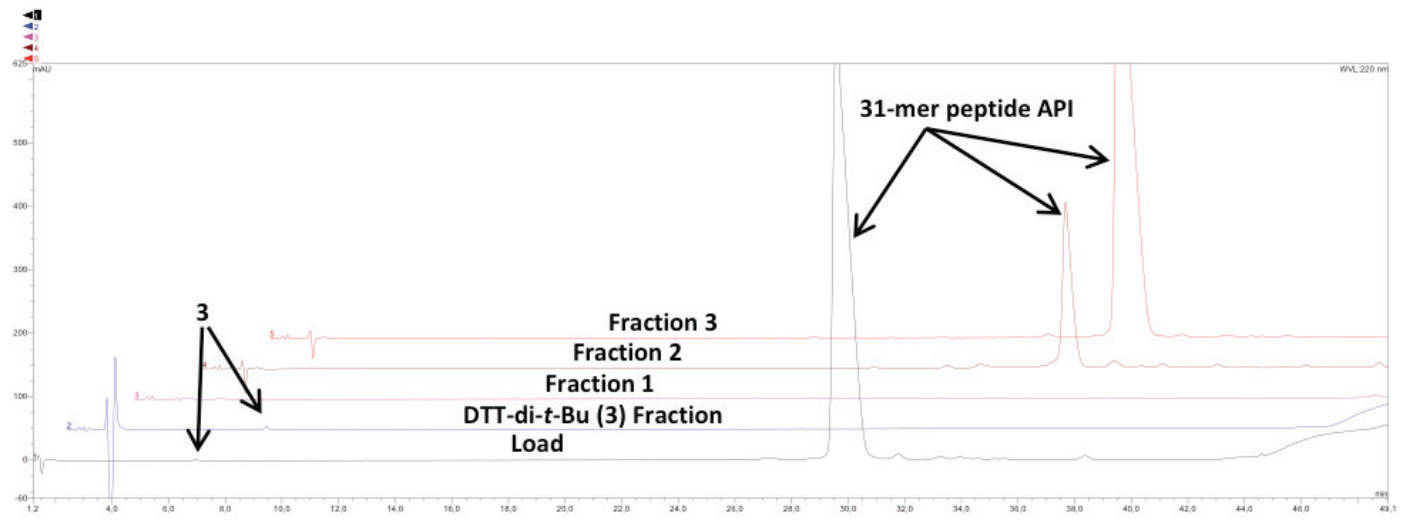
31-mer peptide API
3
Fraction 3
Fraction 2
Fraction 1
DTT-di-t-Bu (3) Fraction
Load

Figure 4. Removal of the adduct **3** from a solution of a 31-mer peptide API by RP-HPLC. Stationary phase: silica C18; mobile phase: 10 mM phosphate buffer pH 7.5/MeCN. Note that this RP-HPLC run was not aimed at increasing the purity of the peptide API.

Summary

*Herein we reported that DTT is not only a powerful reducing agent but it is also a highly efficient t-Bu$^+$ scavenger. During the course of its action DTT forms either its TFA adduct **2** and/or the di-t-Bu adduct **3**, depending on the parameters of the particular TFA cleavage. We have shown that small organic molecules such as these DTT adducts can in fact co-elute with high MW peptides during downstream processing of therapeutic peptides. Our experiences with compounds **2** & **3** illustrate that controlling low MW non-peptide impurities in peptide manufacturing is equally important as controlling their peptide related counterparts.*[11]

References

[1] Correspondence to jap@polypeptide.com.

[2] See for example a) Swietlow, A.; Lax, R. Chimica Oggi/Chem. Today 2004, July/August, 22. b) Zompra, A. A.; Galanis, A. S. Werbitzky, O.; Albericio, F. Future Med. Chem. **2009**, 1, 361.

[3] Cleland, W. W. Biochemistry **1964**, 3, 480.

[4] For example of use of DTT in large scale peptide (Fuzeon) manufacturing see Zhang, H., Schneider, S. E., Bray, B. L., Friedrich, P. E., Tvermoes, N. A., Mader, C. J., Whight, S. H., Niemi, T. E., Silinski, P., Picking, T., Warren, M., Wring, S. A. Org. Process Res. Dev. **2008**, 12, 101.

[5] Pawlas, J.; Hansen, S.; Sørensen, A. H.; Stærkær, G.; Møller, A.; Thompson, N.; Pagano, T.; Kong, F.; Koza, S.; Pozzo, M.; Finneman, J.; Droege, J. In Peptides 2010. Proceedings of the 31st European Peptide Symposium; Lebl, M.; Meldal, M.; Jensen, K. J. Eds.; Prompt Scientific Publishing: San Diego, U.S.A. 2010; pp 132.

[6] M.p. 77-79 °C (EtOH/H_2O). M.p. lit 80 °C, see ref 8.

[7] Anal. Calcd for $C_{12}H_{26}O_2S_2$ (**3**): C, 54.1; H, 9.8; S, 24.1. Found: C, 54.4; H, 9.8; S, 24.5.

[8] ^{1}H NMR spectrum of **3** ($CDCl_3$, 400 MHz) was in keeping with the ^{1}H NMR spectrum of (2R,3R)-1,4-bis(tert-butylthio)butane-2,3-diol (**3**) prepared from tartaric acid. See Ishizaki, M; Hoshino, O. Chirality, **2003**, 15, 300.

[9] ^{13}C NMR ($CDCl_3$, 100 MHz): δ 71.55 (CH), 42.57 (C_q), 32.86 (CH_2), 31.05 (CH_3).

[10] See ref 5 for the details of this experiment.

[11] We thank Ms. Sylvia Ritz for editorial assistance.

www.polypeptide.com